Risk Factors Correlating With MC4R Pathway Variants on Genetic Testing

Patrick Sleiman,[1] Elliot Bromberg,[2] Rick Norton,[1] Jill C. Garrison[1]
[1]Rhythm Pharmaceuticals, Inc., Boston, MA, USA; [2]Beghou Consulting, Inc., Rockville, MD, USA

To download a PDF version of this presentation please scan or visit https://rhythmtx.com/publications/

Introduction

- Hyperphagia, or pathologic uncontrollable hunger, and early-onset, severe obesity can result from rare genetic variants that disrupt key regulatory pathways of hunger and energy expenditure regardless of environmental factors[1]
 - Variants in key genes of the melanocortin-4 receptor (MC4R) pathway, a regulator of energy balance, have been associated with obesity and hyperphagia[1-4]
- The diagnosis of rare genetic obesity diseases may be improved by the expansion of genetic testing, which can allow access to specialized and indicated therapeutic approaches[1,5-8]
 - Currently, low use of genetic testing may contribute to the underdiagnosis and underreporting of rare genetic causes of obesity[2,9]
- The goal of the Uncovering Rare Obesity® (URO) program (www.UncoveringRareObesity.com) is to enhance access to genetic testing for individuals with suspected rare genetic causes of obesity in the United States and Canada[10]

Objective

- To investigate the prevalence of gene variants associated with obesity and their relationship to demographic characteristics, obesity, hyperphagia, and family history of genetic disease if earlier testing results are available

Methods

- Eligible consented individuals were tested per physician order for gene variants in a panel of 79 genes and 1 chromosomal region known to be associated with obesity as part of the URO program
- A positive genetic test result was defined as identifying variant(s) classified by the American College of Medical Genetics as pathogenic or likely pathogenic in a manner that is consistent with the mode of inheritance known for causing disease in the respective gene (ie, biallelic variants in a single gene with autosomal recessive inheritance)
- The Phase 3 EMANATE clinical trial (NCT05093634) is assessing the efficacy and safety of the MC4R agonist setmelanotide in patients with obesity due to leptin receptor insufficiency (*LEPR* heterozygous), proopiomelanocortin insufficiency (*POMC* or *PCSK1* heterozygous), steroid receptor coactivator 1 (SRC1) deficiency, and SH2B adapter protein 1 (SH2B1) deficiency
- Using deidentified data, correlations between detected genetic variants and characteristics of patient obesity, hyperphagia, and family history were assessed using linear regression for continuous variables or chi-square tests for categorical values

Table 1. The Uncovering Rare Obesity® Gene Panel

79-gene and 1 chromosomal region panel				
ADCY3	*CPE*	*LEPR*	*PHF6*	*SEMA3C*
AFF4	*CREBBP*	*LZTFL1 (BBS17)*	*PHIP*	*SEMA3D*
ALMS1	*CUL4B*	*MAGEL2*	*PLXNA1*	*SEMA3E*
ARL6 (BBS3)	*DNMT3A*	*MC3R*	*PLXNA2*	*SEMA3F*
BBIP1 (BBS18)	*DYRK1B*	*MC4R*	*PLXNA3*	*SEMA3G*
BBS1	*EP300*	*MECP2*	*PLXNA4*	*SH2B1*
BBS10	*GNAS*	*MKKS (BBS6)*	*POMC*	*SIM1*
BBS12	*HTR2C*	*MKS1 (BBS13)*	*PPARG*	*TBX3*
BBS2	*IFT172*	*MRAP2*	*PROK2*	*TRIM32 (BBS11)*
BBS4	*IFT27 (BBS19)*	*NCOA1 (SRC1)*	*RAB23*	*TRPC5*
BBS5	*IFT74 (BBS20)*	*NR0B2*	*RAI1*	*TTC8 (BBS8)*
BBS7	*INPP5E*	*NRP1*	*RPGRIP1L*	*TUB*
BBS9 (PTHB1)	*ISL1*	*NRP2*	*RPS6KA3*	*UCP3*
BDNF	*KIDINS220*	*NTRK2*	*SDCCAG8 (BBS16)*	*VPS13B*
C8ORF37 (BBS21)	*KSR2*	*PCNT*	*SEMA3A*	*WDPCP (BBS15)*
CEP290 (BBS14)	*LEP*	*PCSK1*	*SEMA3B*	16p11.2*

*Assessment for rearrangement of the 16p11.2 chromosomal region.

Design of the URO program

- The URO program gene panel includes 79 genes and 1 chromosomal region of clinical relevance (Table 1)
- Individuals within the United States and Canada were eligible to receive a no-charge genetic test and 2 genetic counseling sessions if they met the eligibility criteria in Figure 1

Figure 1. Eligibility criteria for the Uncovering Rare Obesity® program.

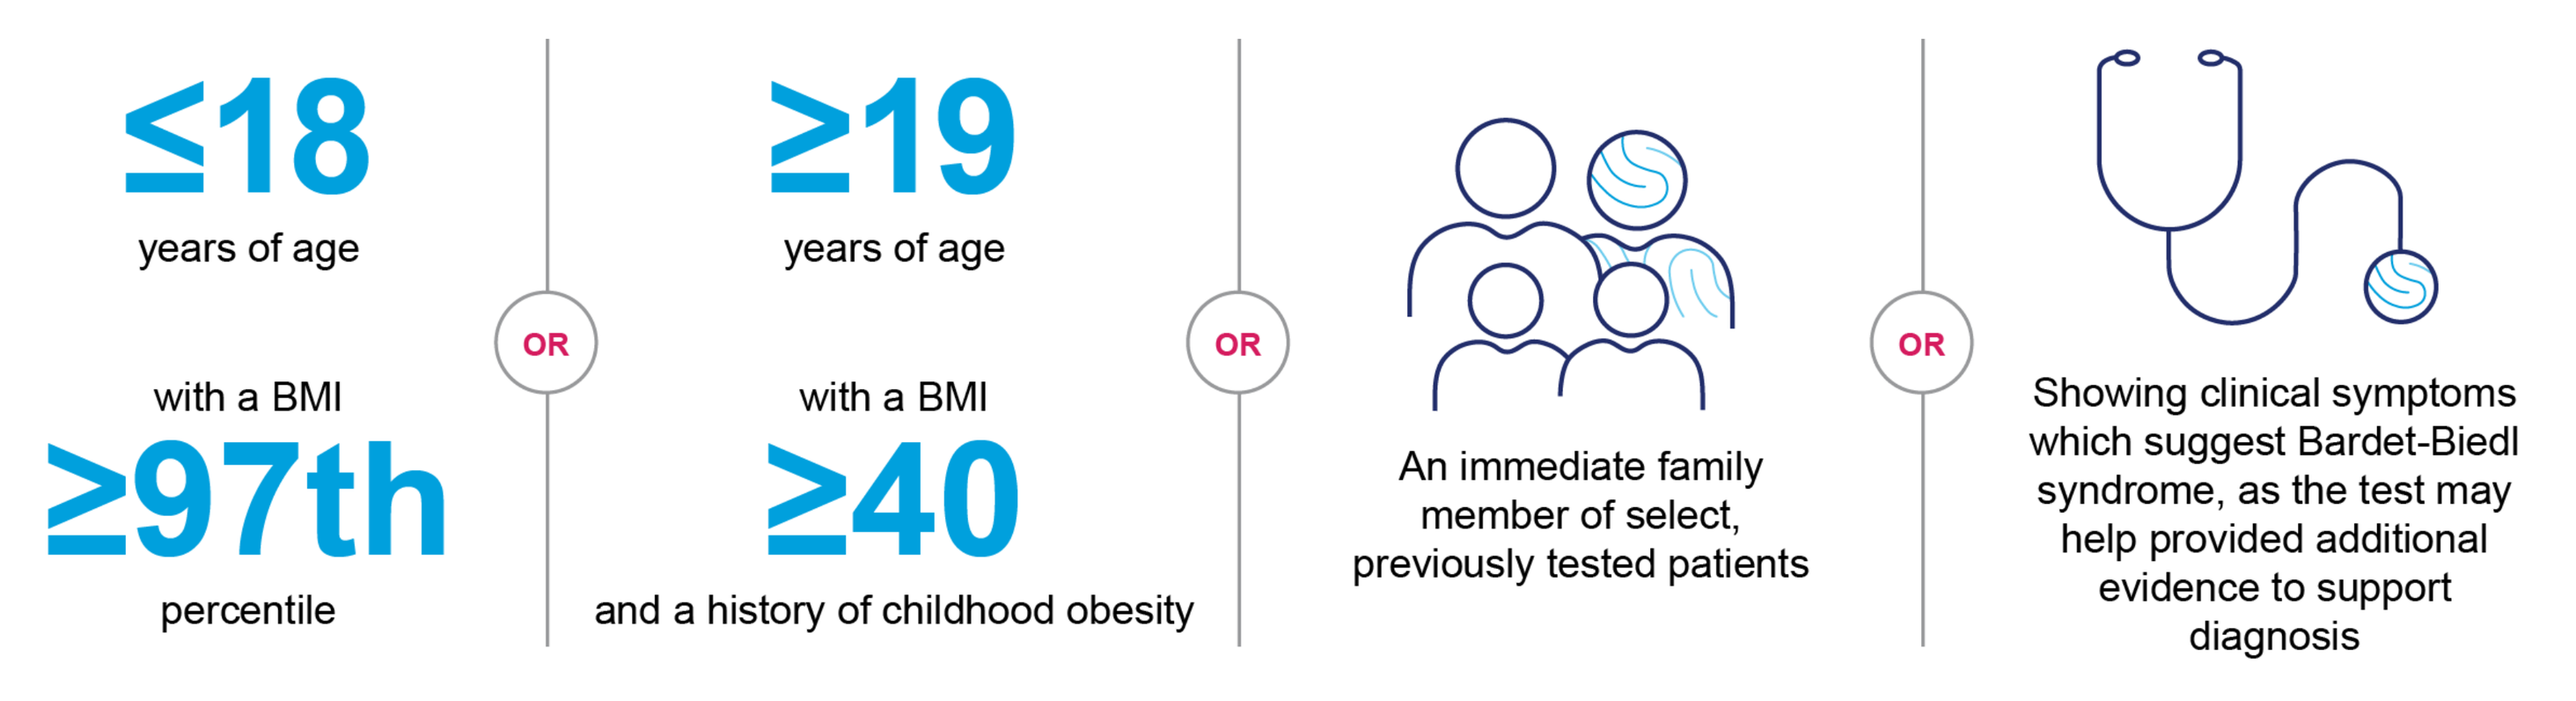
≤18
years of age
with a BMI
≥97th
percentile
OR
≥19
years of age
with a BMI
≥40
and a history of childhood obesity
OR
An immediate family member of select, previously tested patients
OR
Showing clinical symptoms which suggest Bardet-Biedl syndrome, as the test may help provided additional evidence to support diagnosis

BMI, body mass index.

Results

- At the time of this analysis (June 2024), 43,014 individuals had been tested in the URO program
 - Of these individuals, 30,012 (68%) were aged <18 years and 13,002 (32%) were aged ≥18 years
 - Of individuals aged <18 years who participated in the URO program, 49% were male and 51% were female, and of individuals aged ≥18 years, 25% were male and 75% were female
- Overall, 5% of URO participants had a genotype consistent with genetic eligibility for setmelanotide therapy either through approved indications (obesity due to biallelic variants in *POMC*, *PCSK1*, and *LEPR* and Bardet-Biedl syndrome [BBS]) or carried an EMANATE-eligible genotype
- Fifty-three genes with ≥1 causal variant were detected, the most common of which were variants in *MC4R* (n=667; 1.6%), and the most common copy number variant was 16p11.2 deletion duplications (n=108; 0.25%)
- The demographics of individuals with a positive result were generally similar to those of all individuals tested, although notably, the proportion of individuals who were Black or African American was lower for positive individuals (Figure 2)

Figure 2. Race distribution.

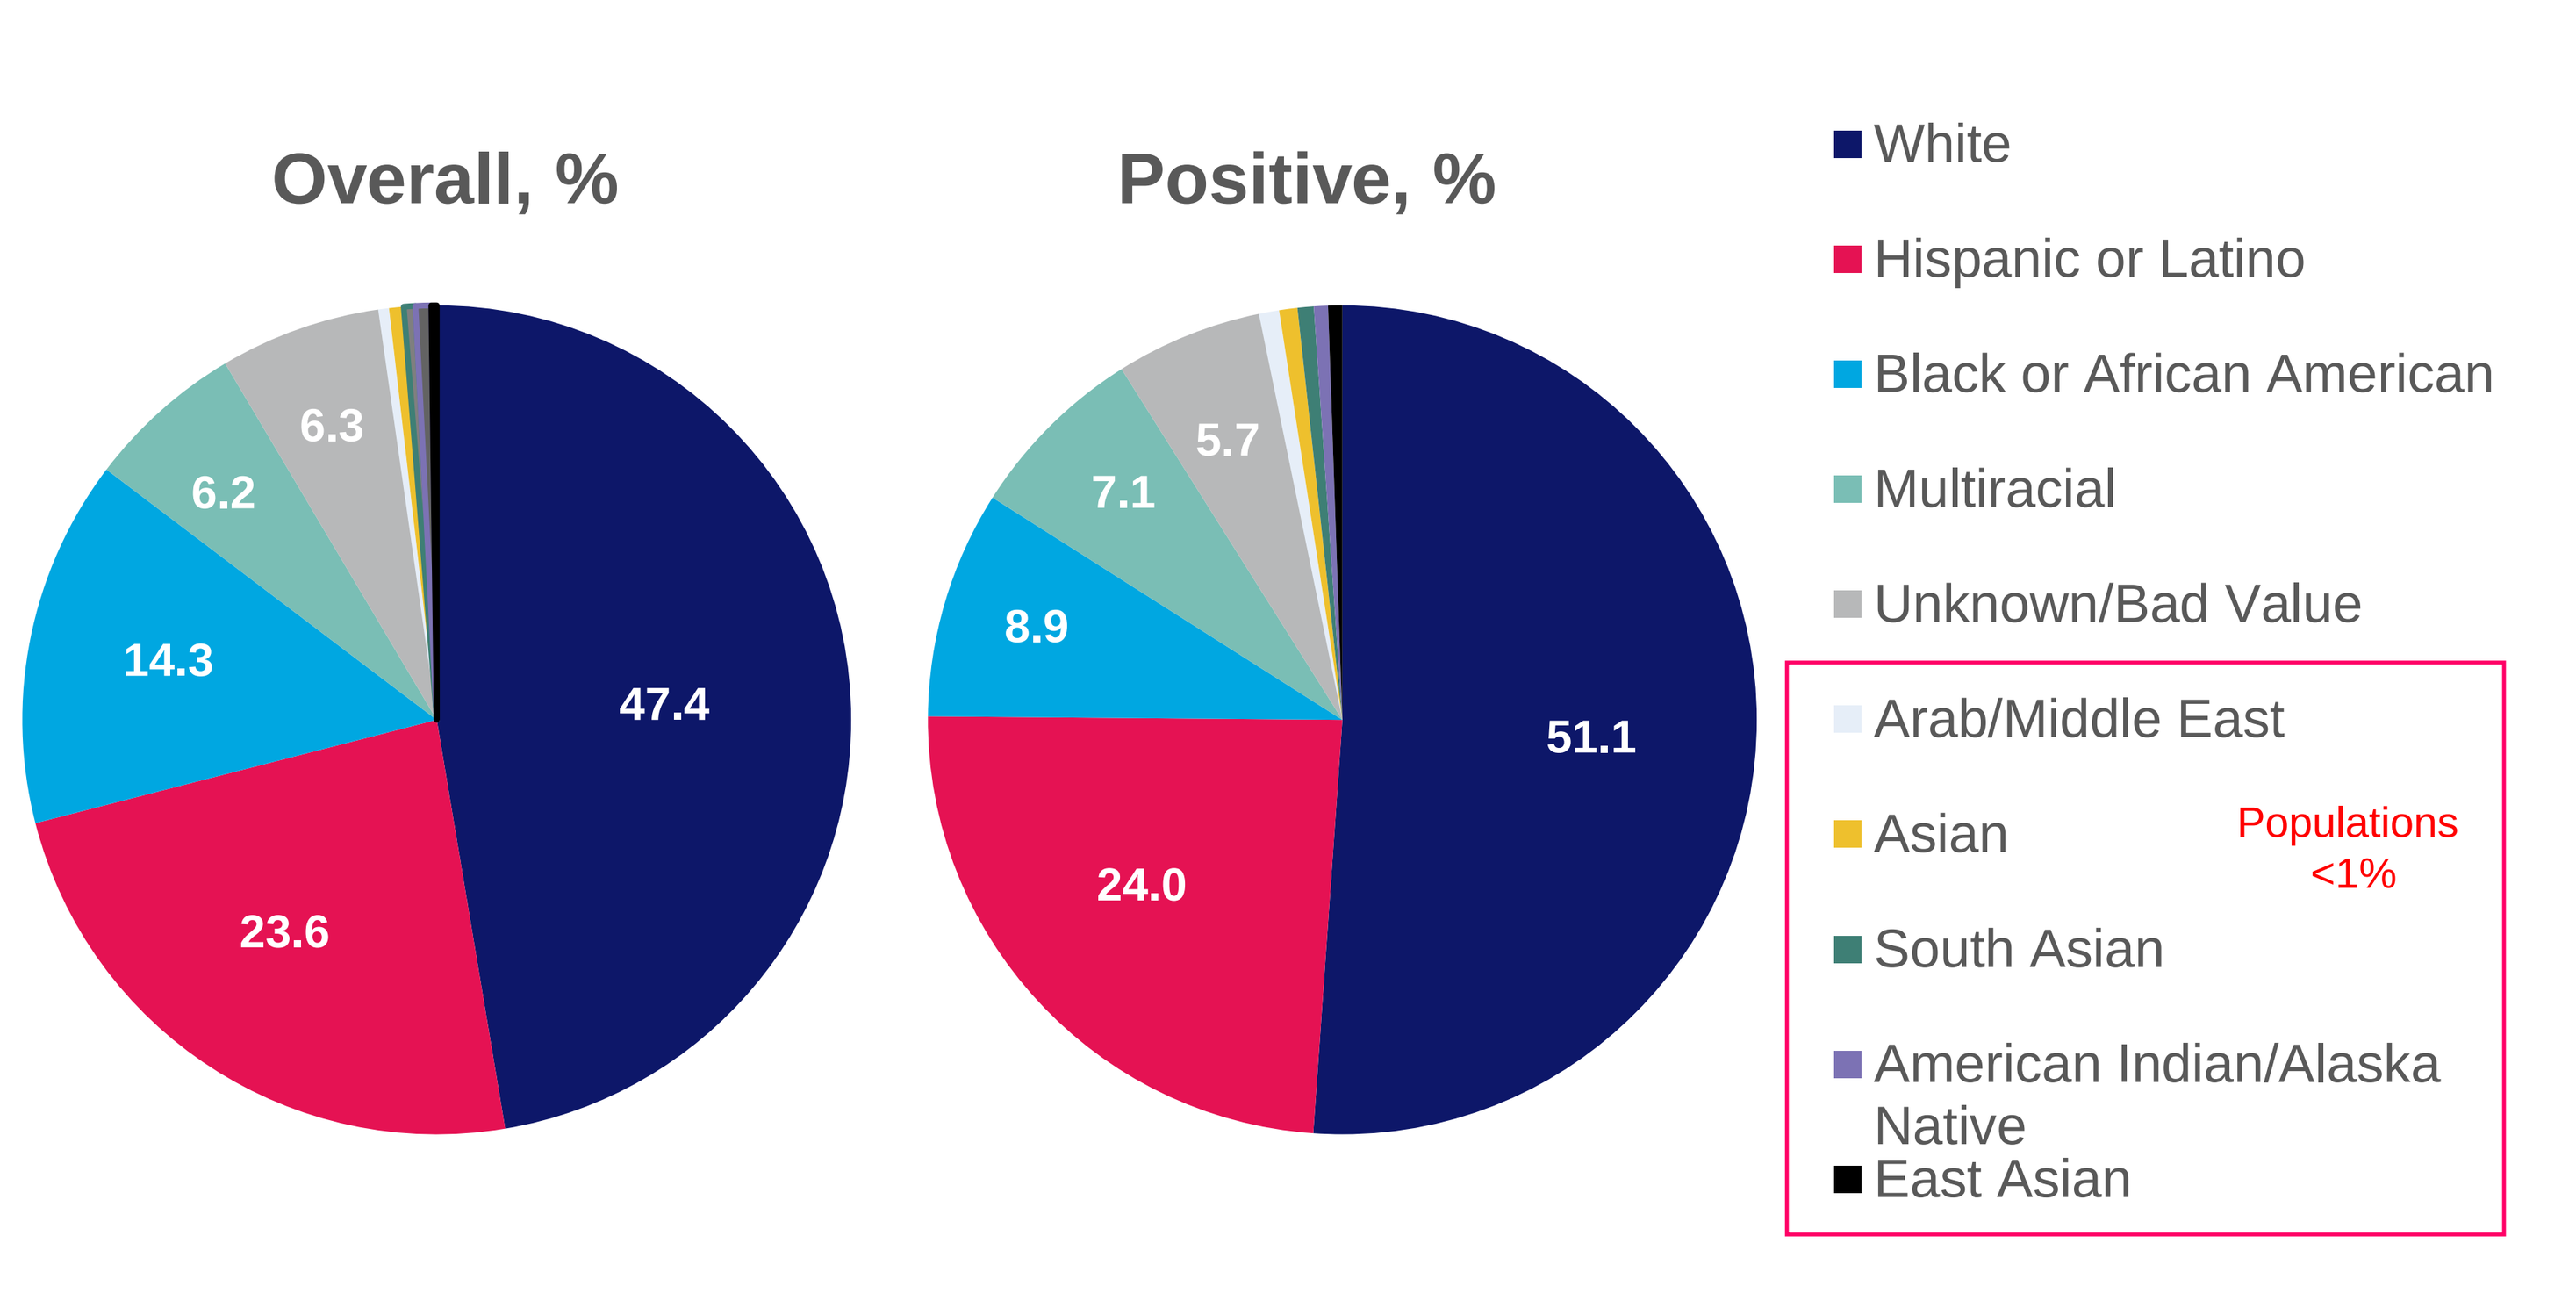
Overall, %
47.4
23.6
14.3
6.2
6.3
Positive, %
51.1
24.0
8.9
7.1
5.7
White
Hispanic or Latino
Black or African American
Multiracial
Unknown/Bad Value
Arab/Middle East
Asian
South Asian
American Indian/Alaska Native
East Asian
Populations <1%

- BBS genetic variants of pathogenic relevance, particularly *BBS1* and *BBS10*, were most commonly reported in White individuals (Table 2)
 - Notably, *BBS10* was not observed in Black or African American individuals and was generally underrepresented in Hispanic or Latino individuals
 - URO pathogenic/likely pathogenic homozygous results (n=51) were found to be enriched in individuals with Hispanic or Latino ethnicity, and this enrichment was found not to be related to geographic location
 - There were an additional 136 cases with biallelic compound heterozygous variants that were of uncertain significance but suspected likely pathogenic or pathogenic
 - Of those 136 indeterminate cases, 5 were biallelic *BBS1* and 9 were biallelic *BBS10* cases

Table 2. Bardet-Biedl Syndrome Genetic Results of Clinical Relevance

Gene	White	Hispanic or Latino	Black or African American	Asian
BBS1	18	14	1	0
BBS10	10	2	0	0
BBS12	0	6	1	0
BBS2	6	0	1	2
BBS4	0	1	0	0
BBS5	0	1	1	1
BBS7	3	1	0	3
BBS9	1	2	2	0
CEP290 (BBS14)	2	2	1	0
MKKS (BBS6)	1	3	0	0
ARL6 (BBS3)	0	2	0	0

- The mean (standard deviation [SD]) age of onset of obesity in individuals who had a positive genetic test result was significantly lower (3.5 [3.5] years) than in those individuals who tested negative (4.8 [3.9] years; *P*<0.0001) (Figure 3)

Figure 3. Age of obesity onset over time.

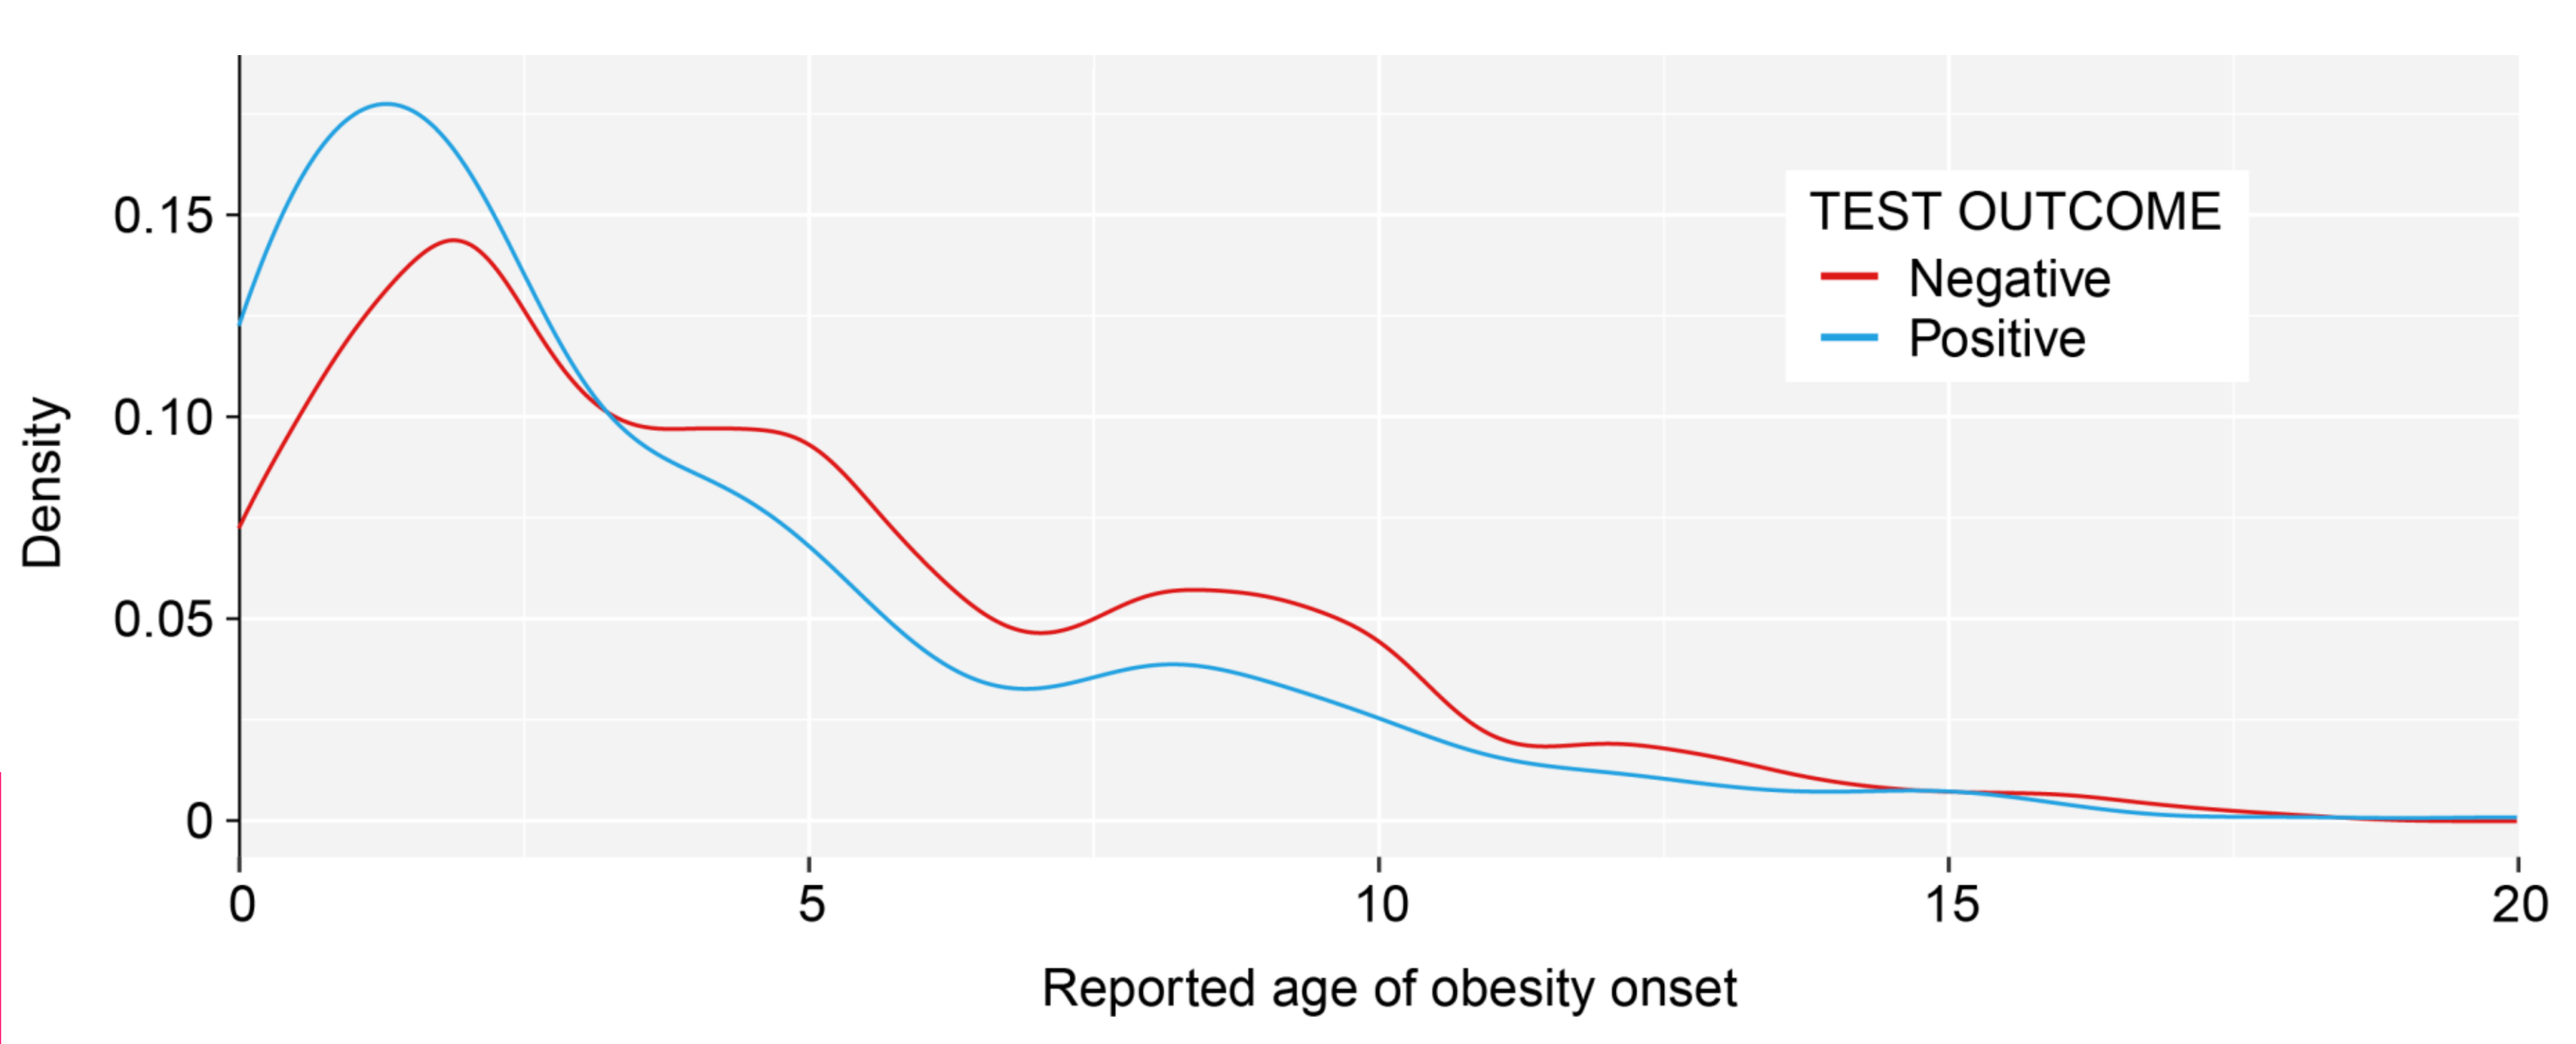
Density
0.15
0.10
0.05
0
0
5
10
15
20
Reported age of obesity onset
TEST OUTCOME
Negative
Positive

- The proportion of individuals with hyperphagia who tested positive (650/1,132; 57%) was significantly higher than in those who tested negative for a variant of interest (6,786/13,708; 49%) (chi-square 2-tailed *P*=0.0010)
- The mean (SD) age of onset of hyperphagia in individuals who were reported to have a positive result was significantly lower (3.4 [3.4] years) than in those individuals who tested negative (4.5 [3.9] years; *P*<0.0001) (Figure 4)

Figure 4. Age of hyperphagia onset over time.

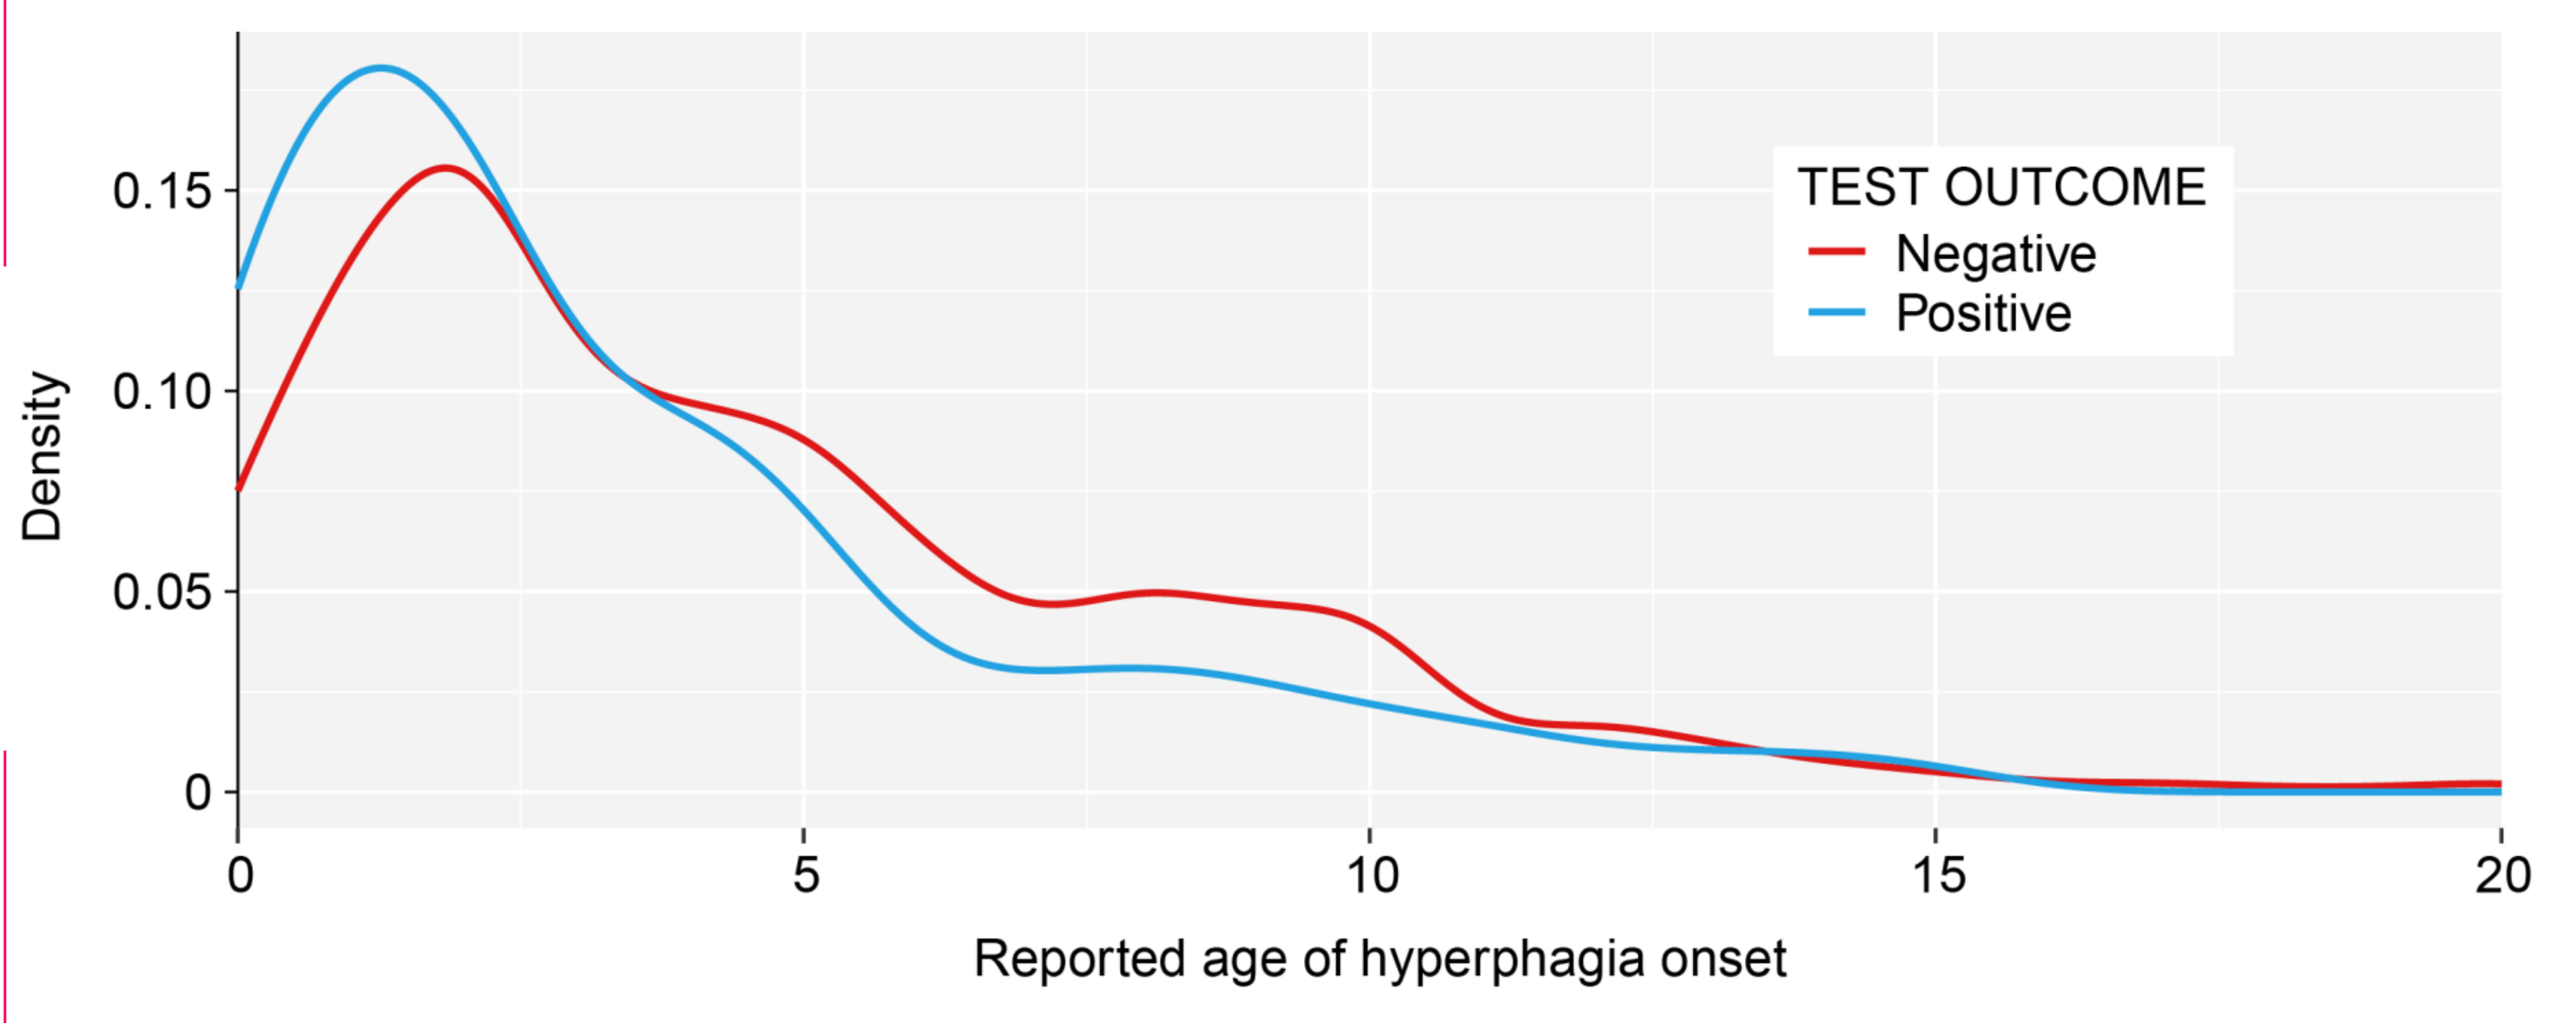
Density
0.15
0.10
0.05
0
0
5
10
15
20
Reported age of hyperphagia onset
TEST OUTCOME
Negative
Positive

- For individuals who tested positive and were reported to have hyperphagia, when stratified by age, the youngest age category (<3 years old) was found to have the highest positive rate (42/650; 6.5%)
- Positive genetic test results indicating an underlying genetic cause of obesity by age are shown in Table 3

Table 3. Positive Genetic Test Results Indicating an Underlying Genetic Cause of Obesity by Age

	0-<3 y	3-<6 y	6-<11 y	11-<18 y	≥18 y	All ages
ACMG positive, %	5.17	3.79	2.66	2.45	2.11	2.63
EMANATE genotype, %	4.95	5.24	4.55	4.82	4.68	4.76
ACMG & EMANATE combined, %	10.12	9.03	7.21	7.27	6.79	7.39

- Family history of genetic disease was also significantly associated with the likelihood of a positive test result (*P*<0.0001), with 15.7% of individuals with a positive test result having a family history of genetic disease versus only 5.3% in individuals with no genetic finding

Conclusions

- The URO results in >40,000 individuals continue to demonstrate the importance of genetic testing in the diagnosis of individuals with a genetic cause underlying their obesity
- Clinical characteristics strongly associated with the positive results include earliest onset obesity and hyperphagia, as well as family history of genetic disease
- Even in a cohort selected for early-onset obesity and the presence of hyperphagia, individuals who tested positive according to ACMG criteria had a mean onset over 1 year earlier than those for whom no genetic cause could be found; onset ranged from birth to peaking at 3 years of age
- Results in the BBS genes demonstrate an ancestry-specific distribution of variants including an enrichment of *BBS1* homozygous individuals of Hispanic or Latino ancestry
- As of July 2024, the URO program gene panel has been expanded to include 7 additional genes related to BBS and the agouti-signaling (ASIP) gene and continues to be available to eligible individuals

Acknowledgments: This study was sponsored by Rhythm Pharmaceuticals, Inc. Writing and editorial support for this poster were provided under the direction of the authors by MedThink SciCom and funded by Rhythm Pharmaceuticals, Inc.

References: 1. Huvenne et al. *Obes Facts*. 2016;9:158-173. **2.** Clément et al. *Physiol Behav*. 2020;227:113134. **3.** Yang et al. *Nat Commun*. 2019;10:1718. **4.** Yazdi et al. *PeerJ*. 2015;3:e856. **5.** Gregoric et al. *Front Endocrinol (Lausanne)*. 2021;12:689387. **6.** Styne et al. *J Clin Endocrinol Metab*. 2017;102:709-757. **7.** van der Valk et al. *Obes Rev*. 2019;20:795-804. **8.** Zorn et al. *Mol Cell Pediatr*. 2020;7:15. **9.** Ayers et al. *J Clin Endocrinol Metab*. 2018;103:2601-2612. **10.** Uncovering Rare Obesity®. Updated July 2024. https://uncoveringrareobesity.com/. Accessed July 31, 2024.

ObesityWeek®; November 3-6, 2024; San Antonio, TX